455PD - First-in-human, first-in-class phase I study of MTL-CEBPA, a RNA oligonucleotide targeting the myeloid transcription factor CEBP-α in patients with advanced hepatocellular cancer (HCC)

MiNA Therapeutics

D. Sarker[1], R. Plummer[2], B. Basu[3], T. Meyer[4], Y.-T. Ma[5], J. Evans[6], D. Palmer[7], K.-W. Huang[8], C.E. Chee[9], D. Spalding[10], M. Sodergren[11], N. Habib[11]

[1]Guy's and St. Thomas' Hospital NHS Trust, London, GB, [2]The Freeman Hospital (NHS Foundation Trust) Northern Centre for Cancer Care, Newcastle upon Tyne, GB, [3]Cambridge University Hospitals NHS Foundation Trust - Addenbrooke's Hospital, Cambridge, GB, [4]UCL Cancer Institute/Paul O'Gorman Building, London, GB, [5]University of Birmingham, Birmingham, GB, [6]Beatson West of Scotland Cancer Centre, Glasgow, GB, [7]Royal Liverpool University Hospital, Liverpool, GB, [8]National Taiwan University Hospital, Taipei, TW, [9]National University Hospital, SG, [10]Imperial College, Hammersmith Hospital, London, GB, [11]Imperial College, Hammersmith Hospital, London, GB

Background

- saRNAs are small oligonucleotide drugs designed to selectively upregulate therapeutic proteins by recruiting endogenous transcription complexes to a target gene, leading to increased expression of naturally processed mRNA
- Transcription factor C/EBP-α (CCAAT/enhancer-binding protein alpha) is a leucine zipper protein which acts as a master regulator of liver homeostasis and multiple oncogenic processes including cell cycle control, proliferation and angiogenesis
- MTL-CEBPA comprises a double stranded RNA payload formulated inside a SMARTICLES® liposomal nanoparticle to specifically target the *CEBPA* gene and has been shown to improve liver function and inhibit HCC tumour growth in preclinical models (Reebye et al, Hepatology, 2014; Voutila et al, Molecular Therapy, 2017; Reebye et al, Oncogene, 2018)
- **MTL-CEBPA is the first saRNA and the first drug targeting C/EBP-α to enter clinical trials**

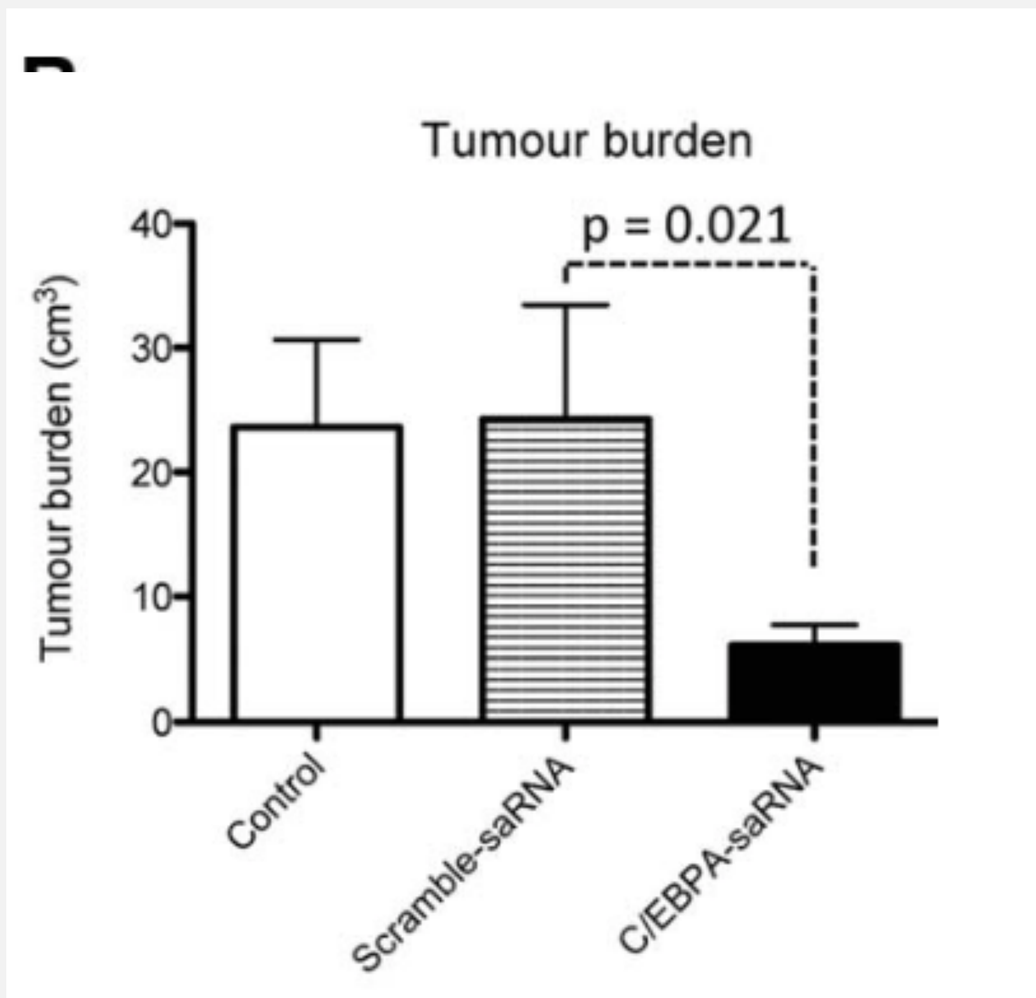
Tumour burden
p = 0.021
Tumour burden (cm³)
40
30
20
10
0
Control
Scramble-saRNA
C/EBPA-saRNA

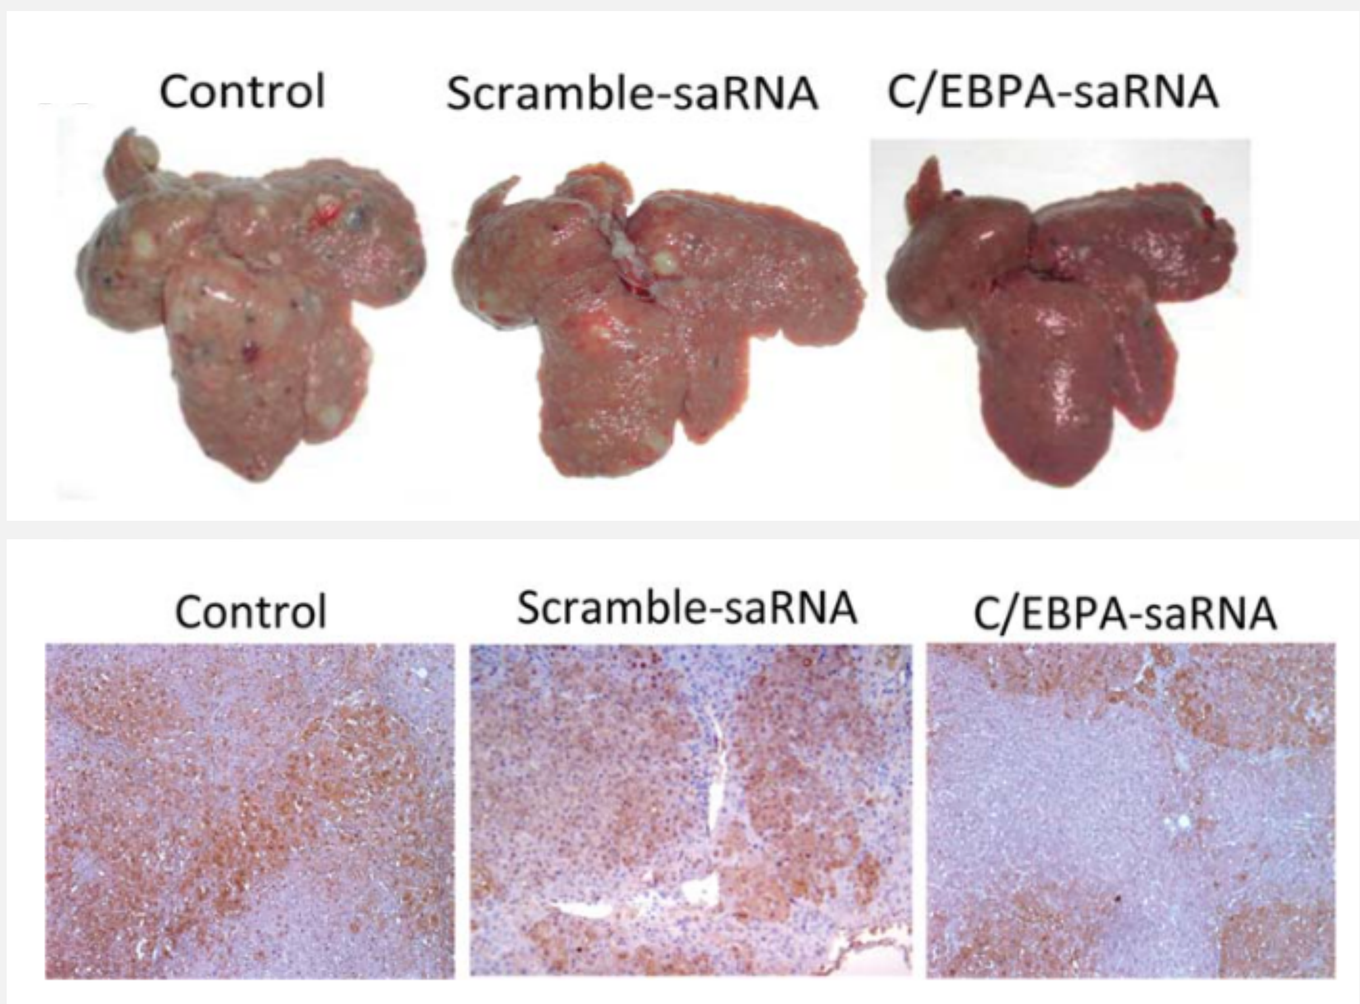
Control
Scramble-saRNA
C/EBPA-saRNA
Control
Scramble-saRNA
C/EBPA-saRNA

Methods

- 4 week cycle (3 weeks dosing + 1 week rest)
- MTL-CEBPA administered by intravenous infusion over 60 minutes (QW/BIW/TIW)
- Pre-medication may be administered to prevent infusion reactions
- AE, PK and PD assessed
- Radiological response determined by RECIST 1.1 every 8 weeks (2 cycles)
- All data are preliminary and based on a data cut-off of May 1, 2019

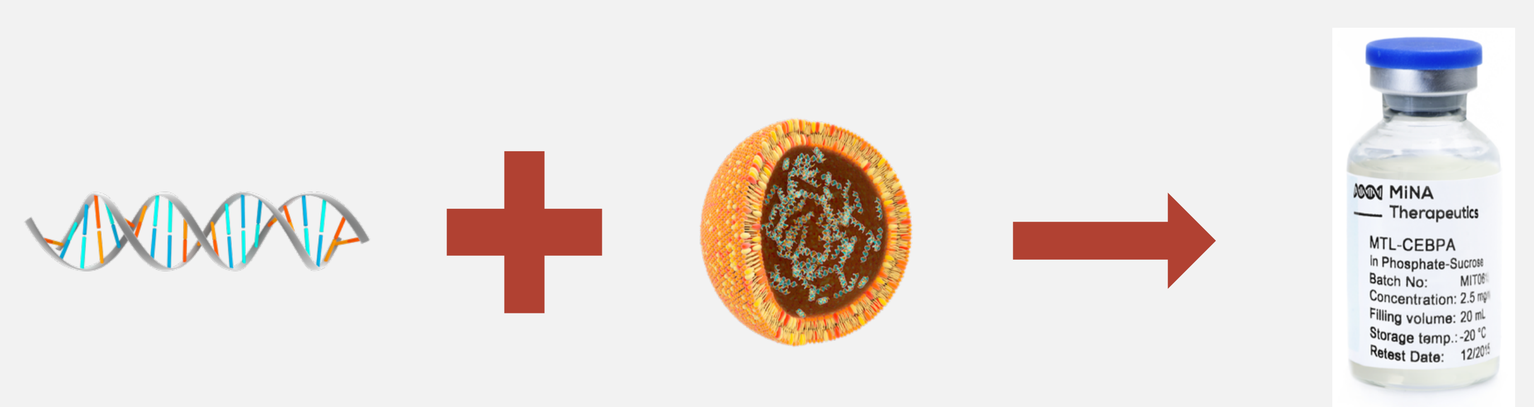
CEBPA-51 saRNA to CEBPA gene
API
NOV340 SMARTICLES® liposome
Formulation
MTL-CEBPA
Drug Product

Study Design

- International multi-centre, open-label, Phase 1 in two parts
- Part 1: 3+3 dose escalation: QW, BIW, TIW
- Part 2: dose expansion at Recommended Phase 2 Dose

Table 1. Dosing

MTL-CEBPA (mg/m²)	No. Recruit. Patient (n=39)
QW Dosing	**25**
28	5
47	4
70	3+3
98	4
130	3
160	3
BIW Dosing	**11**
70 (D1/2)	6
70 (D1/3)	5
TIW Dosing	**3**
70 (D1/2/3)	3

Key Inclusion Criteria

- Unresectable, histologically confirmed HCC or secondary liver cancer
- HCC only recruited from cohort 3± prior sorafenib
- Child-Pugh class A /B with no clinically apparent ascites
- ECOG performance status 0–1
- At least one measurable liver lesion (≥ 1.0cm)
- Life expectancy greater than 3 months at the time of the recruitment
- Platelets ≥ 60 x 10^9/L

Baseline Demographics/Characteristics & Response

- Overall, 39 patients were enrolled with 19 patients (48.7%) completing the first two cycles of treatment
- 35 patients evaluable for efficacy; partial response (PR) was achieved in 1 pt and stable disease (SD) achieved in 15 pts
- In the 28mg/m² QW cohort a 78-year-old female with HCC and cirrhosis on a background of hepatitis B (treated) achieved a confirmed partial response (maintained up to 24 months on treatment) associated with rapid and dramatic decrease of AFP level

Table 2. Efficacy

Best Overall Response	Total (36) / %
Partial Response	1 (2.9)
Stable Disease	15 (42)

Table 3. Demographics

Characteristics, No. (%)	Total (n=39)
Median age, years (range)	65 (27 - 80)
Gender:	
Male	30 (77)
Female	9 (23)
ECOG:	
PS=0	15 (38)
PS=1	23 (59)
PS=2	1 (3)
Tumour Type:	
HCC	34 (87)
Secondary HCC	5 (13)

Safety

- A maximum tolerated dose was not reached
- Most treatment-emergent adverse events (TEAEs) in all cohorts were Grade 1 or 2; Grade 3 TEAEs were reported in 61.5% of patients (24 patients) overall
- Changes in liver function tests were generally transient such that overall there were no significant changes in LFTs at the end of the first and second cycles of treatment compared to baseline
- Sixteen serious adverse events (SAEs) reported in 14 patients (no SAE was reported in more than 1 patient)
- AEs (all SAEs) leading to treatment withdrawal occurred in 3 patients, including acute coronary syndrome, hyperbilirubinemia, and stoma site hemorrhage

Table 4. Treatment-emergent adverse events

Most Frequent TEAEs ≥20% Pts	% Pts
Fatigue	33.3
Diarrhoea	30.8
Anemia	23.1
Vomiting	20.5
Increased ALP	20.5
Increased AST	20.5
Thrombocytopenia	20.5

Post-Treatment TKIs

- After discontinuation of MTL-CEBPA, 8 HCC patients were treated with tyrosine kinase inhibitors (TKI)
- 3/8 TKI naïve-patients had sustained complete response (all 3 patients have survived more than a year without any evidence of tumour recurrence), 1/8 had PR, and 2/8 SD by RECIST 1.1
- Complete responses:
 - 61-year-old male with hepatitis B-related cirrhosis and HCC who was previously treated with ablative therapy, TACE, and doxorubicin
 - 67-year-old male with HCC related to hepatitis C, previously treated with TACE and durvalumab
 - 61-year-old male with hepatitis B and C previously treated with ablative therapy, TACE, and doxorubicin

Pharmacokinetics

- Mean plasma terminal half-life reasonably consistent across dose cohorts after once-weekly treatment with MTL-CEBPA, total plasma clearance and the apparent volume of distribution decrease with increasing dose (see table below)
- Plasma CEBPA concentration at 24h after each dose consistent across all the dosing regimens, showing little accumulation even when dosed once-daily (D1, D2, D3) at 70 mg/m²
- No effect of age (range 27-80 yrs), gender (9F/30M) and concomitant medication on the pharmacokinetics of MTL-CEBPA

Fig 1. Mean plasma terminal half-life

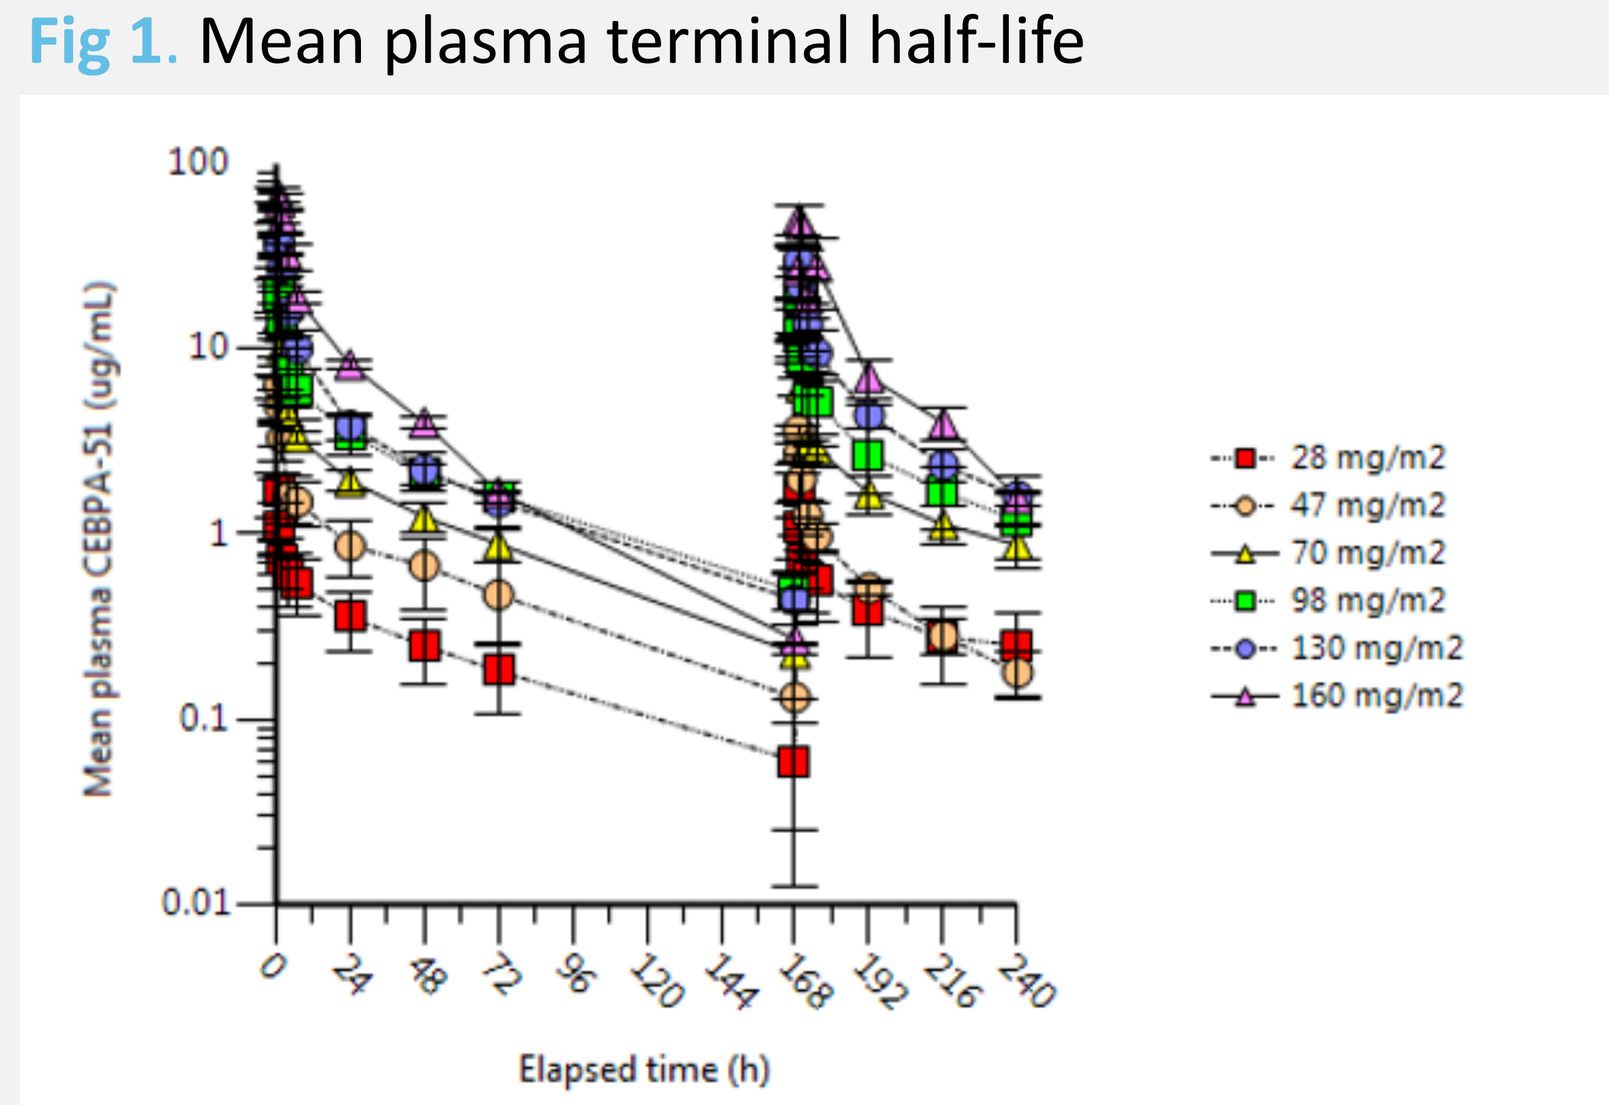
Mean plasma CEBPA-51 (ug/mL)
100
10
1
0.1
0.01
0
24
48
72
96
120
144
168
192
216
240
Elapsed time (h)
28 mg/m2
47 mg/m2
70 mg/m2
98 mg/m2
130 mg/m2
160 mg/m2

Table 5. CEBPA-51 PK parameters – Q1wk cohorts 0-7 days

Dose	n=	Tmax (h)	Cmax (µg/mL)	AUCinf (h.µg/mL)	CL (L/h/M²)	Vz (L/m²)	Half-life (h)
28	5	1.00	1.68 ± 0.98	30.7 ± 30.6	2.08 ± 2.18	98.7 ± 103.0	49.2 ± 20.4
47	4	1.00	6.33 ± 1.87	98.7 ± 106.1	0.91 ± 0.61	37.1 ± 17.6	45.2 ± 20.6
70	6	1.00	15.50 ± 1.51	219.5 ± 82.4	0.37 ± 0.18	21.3 ± 4.1	49.1 ± 20.2
98	3	1.25	20.14 ± 7.62	408.2 ± 113.8	0.26 ± 0.85	21.6 ± 7.9	59.2 ± 18.5
130	3	1.25	39.43 ± 14.24	499.1 ± 186.2	0.29 ± 0.12	20.6 ± 7.9	50.7 ± 12.0
160	3	1.00	67.45 ± 10.92	836.3 ± 51.2	0.19 ± 0.01	8.5 ± 0.7	30.8 ± 4.1

Mean compartmental PK parameters ± SD: Tmax-time if maximum observed concentration (median); Cmax-maximum observed concentration; AUCinf-are under curve calculated from 0-last observed concentration and extrapolated to infinity; CL-total plasma clearance (dose/AUCinf); Vz-apparent volume of distribution based on the terminal phase; half-life-terminal half-life calculated using at least three data points in the terminal phase

Pharmacodynamics

- CEBPA mRNA levels increased by 1.5-fold consistently across all cohorts
- MTL-CEBPA caused significant increases in WBC consistent with CEBP-α dependent granulopoiesis, and decreased expression of adenosine, PD-1, and CXCR4 mRNA

Fig 2. CEBPA mRNA expression in WBCs (n=10)

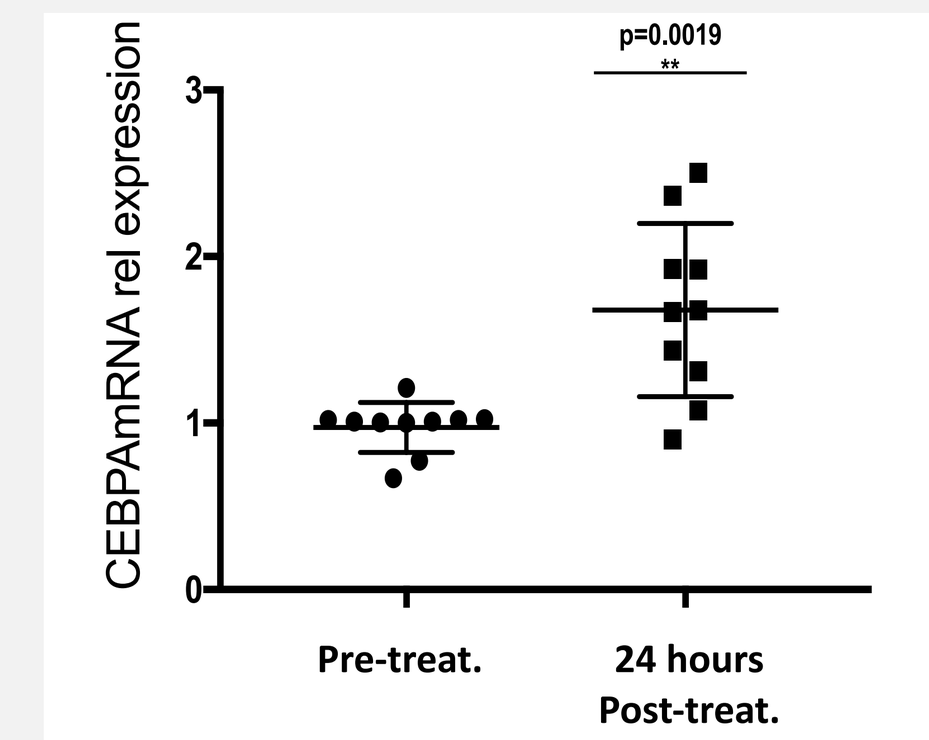
p=0.0019
**
CEBPAmRNA rel expression
3
2
1
0
Pre-treat.
24 hours Post-treat.

Fig 3. WBC vs baseline following dosing at 70 mg/m2 QW (n=5)

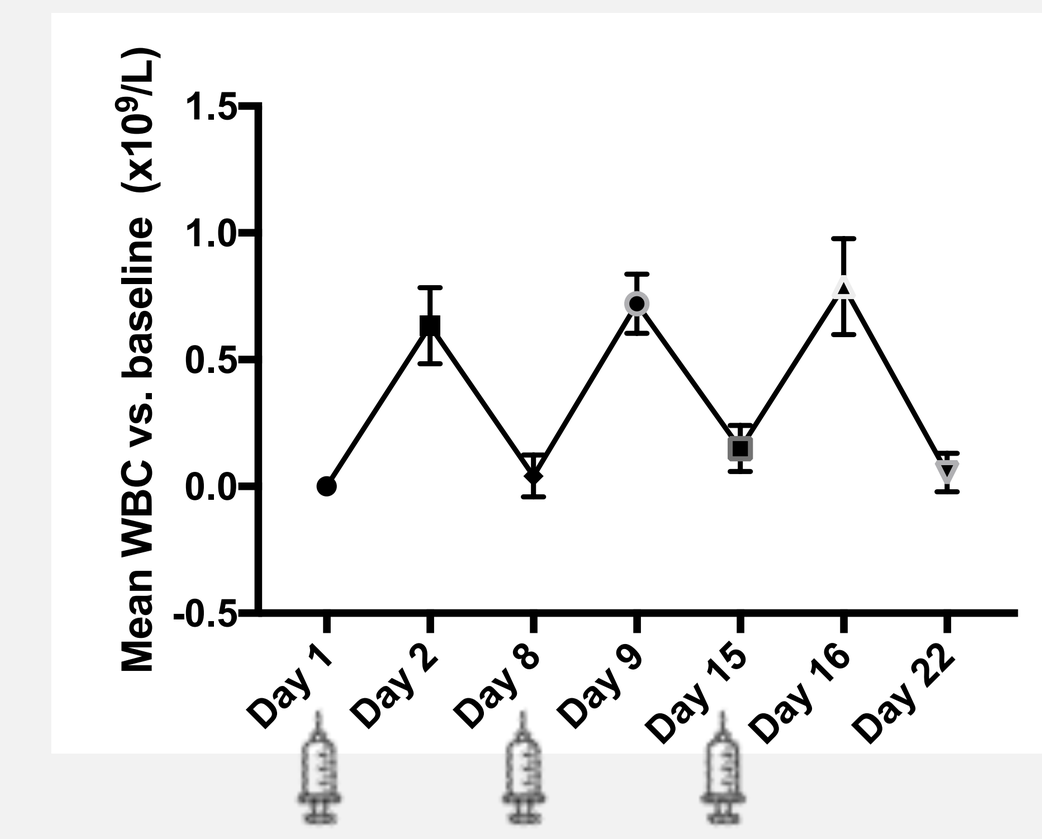
Mean WBC vs. baseline (x10⁹/L)
1.5
1.0
0.5
0.0
-0.5
Day 1
Day 2
Day 8
Day 9
Day 15
Day 16
Day 22

Case Study

Fig 4. Treatment with MTL-CEBPA

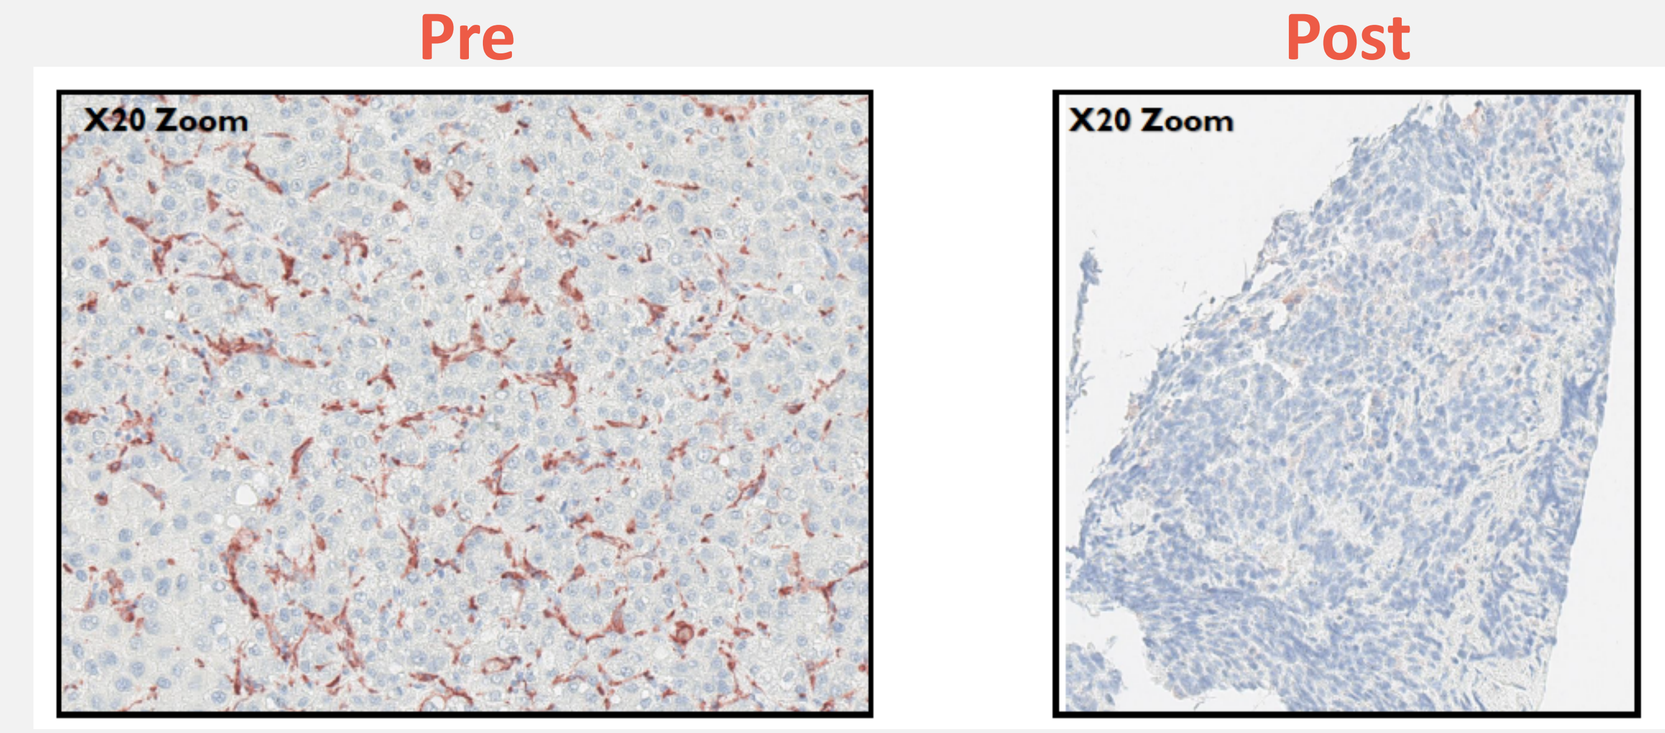
Pre
Post
X20 Zoom
X20 Zoom

- Hypothesized mechanism of action: upregulation of CEBPA gene in myeloid cells leads to repopulation of tumour microenvironment from immuno-suppressive to pathogen-patrolling phenotype
- Two cycles of MTL-CEBPA induced marked reduction of pro-tumour / immuno-suppressive CD163+ M2 macrophages in HCC patient

Fig 5. Treatment with Sorafenib

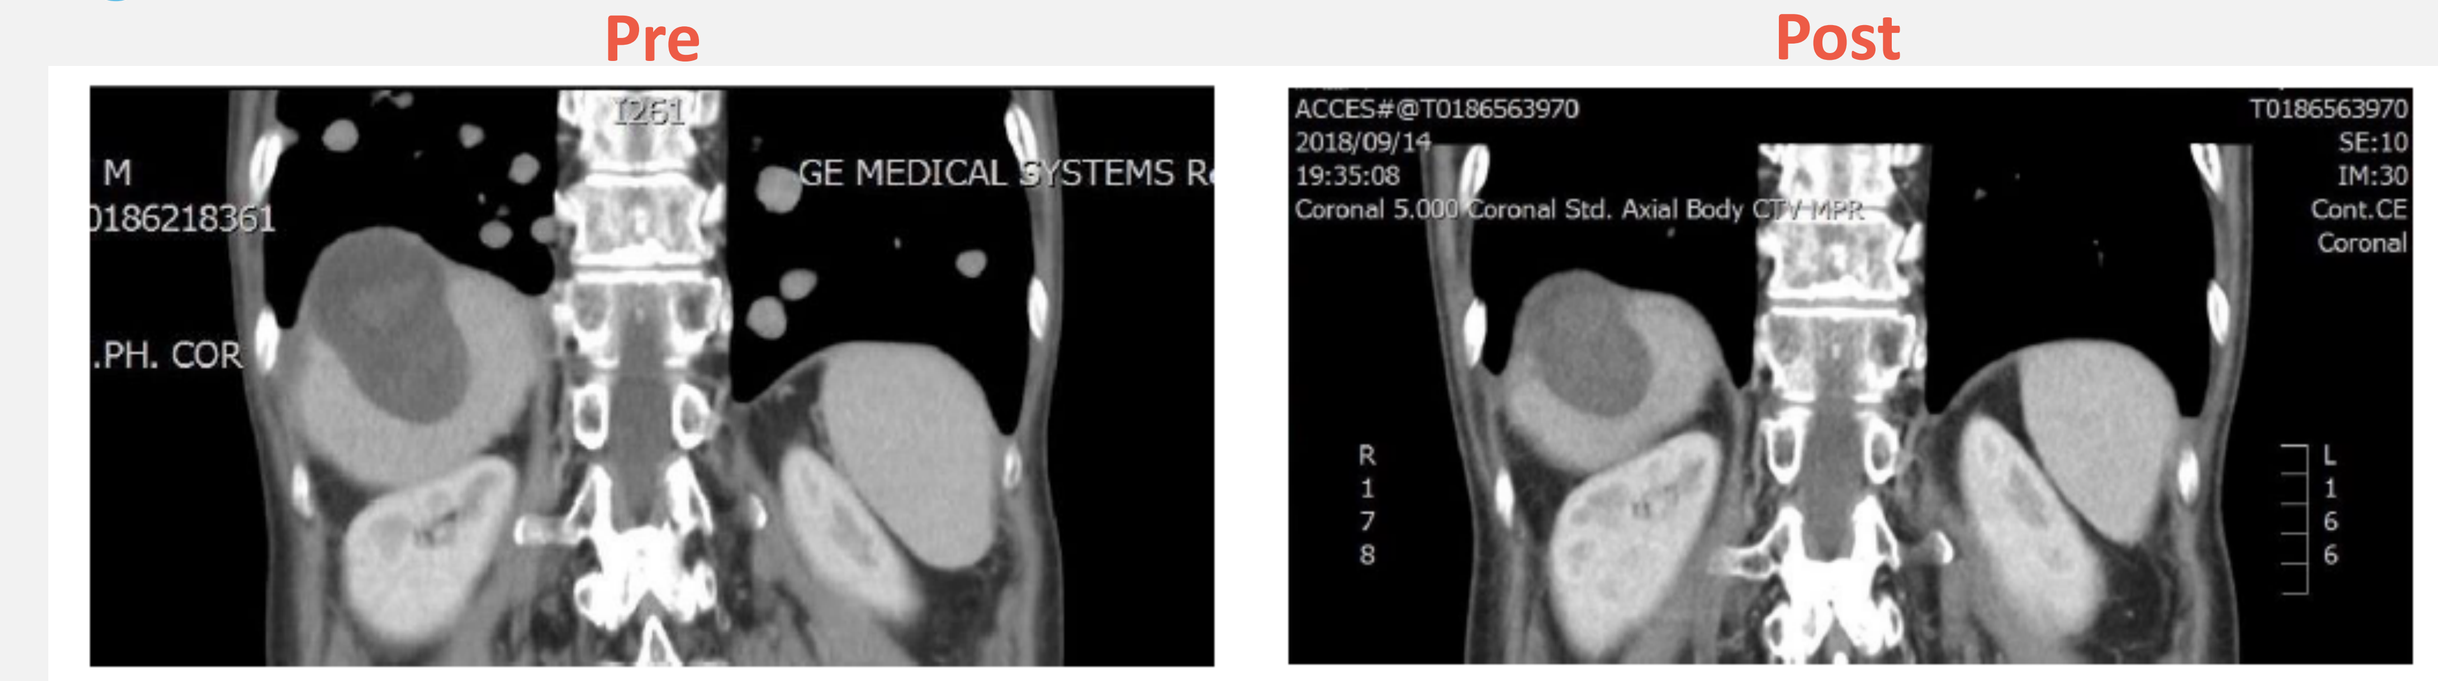
Pre
Post
GE MEDICAL SYSTEMS
ACCES#@T0186563970
2018/09/14
19:35:08
Coronal 5.000 Coronal Std. Axial Body

- 61-year-old male with hepatitis B and C previously treated with ablative therapy, TACE, and doxorubicin who progressed after 2 cycles of MTL-CEBPA (during which time he developed lung metastases)
- Complete radiological response to both lung and liver lesions one month after initiation of TKI treatment
- CT images of the lung lesions are shown above

Discussion

- MTL-CEBPA is well tolerated with no MTD reached; recommended dose for further evaluation is 130mg/m² QW based on safety, PK, and PD
- Encouraging signs of clinical activity are demonstrated, including three complete radiological responses and one partial response in patients treated with MTL-CEBPA and then TKIs
- Hypothesise that MTL-CEBPA sensitises tumours to cancer treatments by repopulating tumour microenvironment from immuno-suppressive to pathogen patrolling myeloid cells
- Encouraging First-in-Human Phase I data validate targeting of C/EBP-α and have prompted further investigation of MTL-CEBPA as combination therapy, including MTL-CEBPA + sorafenib in HCC (recruitment ongoing) and with pembrolizumab (HCC and solid tumours; abstract no 3089)

Acknowledgements: The UK sites receive support from Cancer Research UK and Department of Health as Experimental Cancer Medicine Centres. Financial support for the study was also provided by NIHR Biomedical Research Centre awards.
The study is sponsored by MiNA Alpha Ltd. We would like to thank the participating patients and their families for volunteering to take part in the study, all co-investigators, clinical and nursing staff for the conduct of the study. We also thank Robert Nutbrown and Joanna Nicholls who contributed to this poster.

References
1. Reebye V et al. A novel RNA oligonucleotide improves liver function and inhibits liver carcinogenesis in vivo. Hepatology 2014;59:216-227; 2. Voutila J, et al. Development and Mechanism of Small Activating RNA Targeting CEBPA, a Novel Therapeutic in Clinical Trials for Liver Cancer. Mol Ther. 2017 Dec 6;25(12):2705-2714; 3. Zhao X, et al. Treatment of Liver Cancer by C/EBPA saRNA. Adv Exp Med Biol. 2017;983:189-194; 4. Reebye V et al. Gene activation of CEBPA using saRNA: preclinical studies of the first in human saRNA drug candidate for liver cancer. Oncogene. 2018 Mar 7. doi: 10.1038/s41388-018-0126-2.

Study References: Clinicaltrials.gov: NCT02716012; UK NIHR CRN ID:20332 (CANC 4818)
Contact (Authors): Prof N Habib (email: nagy@minatx.com) and Dr D Sarker (email: debashis.sarker@kcl.ac.uk)
Potential Conflicts of Interest: Prof N Habib is Chief Medical Officer of MiNA Alpha Ltd; Dr D Sarker received travel/accommodation expenses from MiNA Alpha Ltd (to cover attendance at the ESMO meeting)