A phase 1 study of saRNA myeloid modulating agent MTL-CEBPA in combination with pembrolizumab in adult patients with advanced solid tumours

MiNA Therapeutics

Ruth Plummer[1], Mikael H Sodergren[2], David James Pinato[2], Debashis Sarker[3], Vikash Reebye[2,4], Duncan Spalding[2], Nina Raulf[4], Laura Sinigaglia[4], Thomas Talbot[2], Alessio Cortellini[2], Antonio D'Alessio[2], Ilian Tchakov[4], Robert Habib[4], John Rossi[5], Nagy Habib[2,4]

[1]Northern Institute for Cancer Research, Newcastle University, Newcastle upon Tyne, United Kingdom, [2]Department of Surgery & Cancer, Imperial College London, London, United Kingdom , [3]King's College London, London, United Kingdom, [4]MiNA Therapeutics Limited, London, United Kingdom, [5]Department of Molecular and Cellular Biology, Beckman Research Institute of City of Hope, Duarte, CA 91010, USA

Background

MTL-CEBPA is a novel immunotherapy targeting the myeloid cell lineage which has shown promising clinical activity as monotherapy and combination therapy with tyrosine kinase inhibitors in hepatocellular carcinoma (HCC). Immunosuppressive myeloid cells are associated with worse outcomes to checkpoint inhibitors. Pre-clinical data have shown that MTL-CEBPA potentiates the oncological effect of PD-1 inhibitors (Figure 1A below). Further, following reprogramming by MTL-CEBPA, co-culturing MDSCs with T cells leads to enhanced T cell proliferation (Figure 1B below)

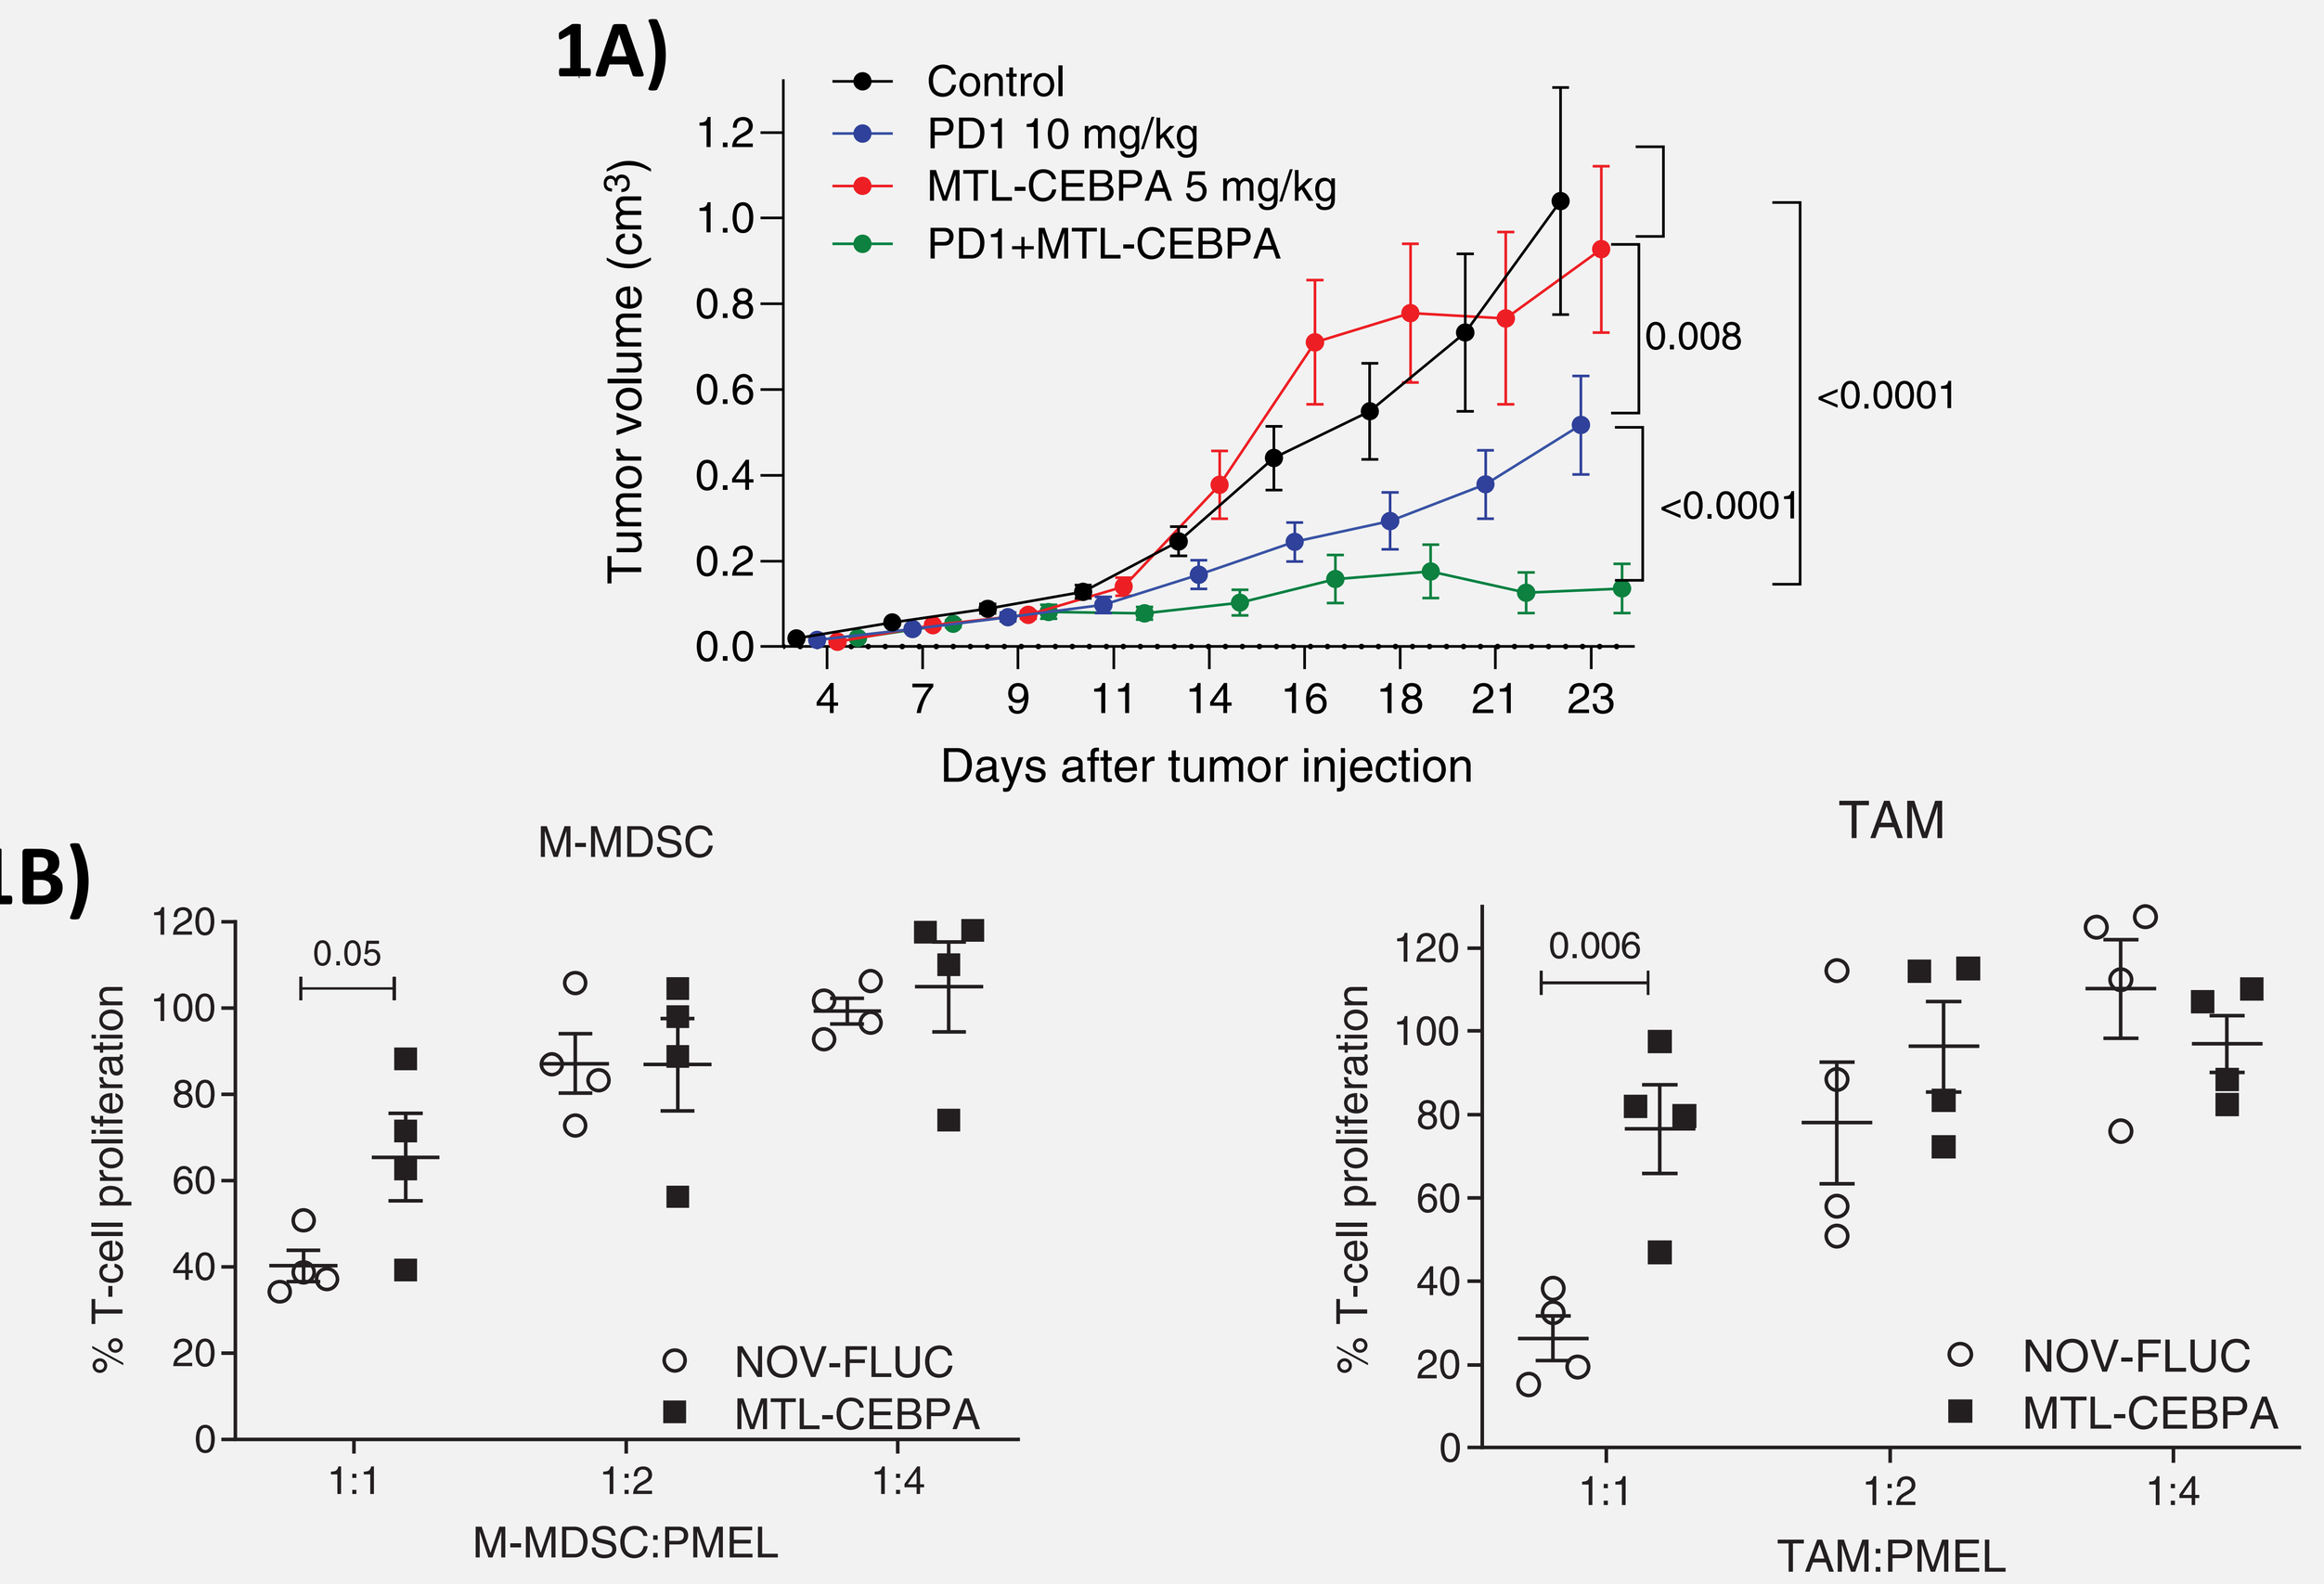
1A)
Control
PD1 10 mg/kg
MTL-CEBPA 5 mg/kg
PD1+MTL-CEBPA
Tumor volume (cm³)
Days after tumor injection
0.008
<0.0001
<0.0001
1B)
M-MDSC
TAM
% T-cell proliferation
0.05
0.006
NOV-FLUC
MTL-CEBPA
M-MDSC:PMEL
TAM:PMEL

Figure 1: MTL-CEBPA enhances the anti-tumour effect of anti-PD-1 treatment in MC38 mice (A). Co-culture of either M-MDSCs or TMAs treated with with MTL-CEBPA significantly increases T cell proliferation (Hashimoto *et al.*, 2021 Clin Can Res).

MTL-CEBPA comprises SMARTICLES® liposomal nanoparticle encapsulating CEBPA-51, a 21-mer small activating 2'O-Me RNA oligonucleotide duplex designed to specifically target and up-regulate transcription of the CEBPA gene

Methods

This phase 1A/B, first-in-human, open-label, multicenter study evaluates the safety, tolerability, PK, and efficacy of MTL-CEBPA in combination with a pembrolizumab in adult patients with advanced solid tumours across 3 dose cohorts (70mg/98mg/130mg/m2 MTL-CEBPA via IV infusion once weekly for 3 consecutive weeks with final week break per cycle, with 200mg pembrolizumab every 3 weeks) between November 2019 and 7 April 2021. This study comprises a dose escalation of 3 planned cohorts in a 3 + 3 design. The primary endpoint is safety (1A) and ORR (RECIST1.1) (1B); key secondary endpoints include PK, CR rate & DCR. Key inclusion criteria: Patients with advanced solid tumours who have progressed on standard of care therapy or for whom no standard therapy is available, measurable disease, ECOG PS <2, life expectancy >3 months. A dose exploration will determine the maximum tolerated dose (MTD) or recommended phase 2 dose (RP2D). MTL-CEBPA dosing was preceded by prednisolone/hydrocortisone and anti-histamine administration to minimise the risk of infusion reactions.

Results

Patient Demographics

	QW 70 mg/m2 N=4	QW 98 mg/m2 N=3	QW 130 mg/m2 N=3	Total N=10
Age (years)	44.5 (44.5)	49.7 (56.0)	49.3 (48.0)	47.5 (50.5)
Sex, male	25%	33%	33%	30%
Race				
Asian or Asian British	0	67%	33%	30%
White	100%	33%	67%	70%
Primary tumour				
Breast cancer	25%	0	0	10%
Cholangiocarcinoma	0	33%	0	10%
Colorectal cancer	25%	0	0	10%
Eccrine carcinoma	0	0	33%	10%
Epithelioid mesothelioma	25%	0	0	10%
Hepatocellular carcinoma	0	33%	0	10%
Ovarian cancer	25%	0	33%	20%
Pancreatic carcinoma	0	0	33%	10%
Thymic cancer metastatic	0	33%	0	10%
ECOG, 0 I 1	75% I 25%	67% I 33%	33% I 67%	60% 40%
Mean/median prior lines of anti-cancer systemic therapy	4.3 (4.5)	1.5 (1.5)	3.0 (2.0)	3.2 (2.0)

Prior Lines of Anti-cancer Treatment

	QW 70 mg/m2 N=4	QW 98 mg/m2 N=3	QW 130 mg/m2 N=3	Total N=10
Prior lines of systemic anti-cancer therapy				
Missing	0	33%	0	10%
1	0	33%	33%	20%
2	25%	33%	33%	30%
4	25%	0	0	10%
5	25%	0	0	10%
6	25%	0	33%	20%

Overview of Adverse Events

Description	QW 70 mg/m2 (N=4)	QW 98 mg/m2 (N=3)	QW 130 mg/m2 (N=3)	All (N=10)
Any TEAE	4 (100%)	3 (100%)	3 (100%)	10 (100%)
- Treatment-Related*	4 (100%)	3 (100%)	3 (100%)	10 (100%)
- MTL-CEBPA Related Only	3 (75%)	1 (33%)	3 (100%)	7 (70%)
- Pembrolizumab Related Only	2 (50%)	2 (67%)	1 (33%)	5 (50%)
Any TEAE Grade 3 or Higher	2 (50%)	0	1 (33%)	3 (30%)
- Treatment-Related*	2 (50%)	0	0	2 (20%)
- MTL-CEBPA Related Only	0	0	0	0
- Pembrolizumab Related Only	1 (25%)	0	0	1 (10%)

TEAEs – AEs related to MTL-CEBPA only

Treatment Emergent Adverse Events	QW 70 mg/m2 N=4		QW 98 mg/m2 N=3		QW 130 mg/m2 N=3		Total N=10	
MTL-CEBPA Related Only, n (%)	Any grade	>=3	Any grade	>=3	Any grade	>=3	Any grade	>=3
Nausea	2 (50)	0	1 (33)	0	0	0	3 (30)	0
Anaemia	0	0	0	0	1 (33)	0	1 (10)	0
Headache	1 (25)	0	0	0	0	0	1 (10)	0
Insomnia	0	0	0	0	1 (33)	0	1 (10)	0
Neutropenia	1 (25)	0	0	0	0	0	1 (10)	0
Pyrexia	0	0	0	0	1 (33)	0	1 (10)	0
Transaminases increased	0	0	0	0	1 (33)	0	1 (10)	0
Ventricular extrasystoles	0	0	0	0	1 (33)	0	1 (10)	0

Safety Results Summary

- No Dose-Limiting Toxicity observed
- No overlapping or potentiating toxicity
- Common (>2 patients) TEAEs observed were diarrhoea, fatigue, AST, ALT and AP increases, nausea, arthralgia, headache
- No AEs leading to dose interruptions and no AEs leading to discontinuation related to MTL-CEBPA treatment
- No CTC Gr3 or worse, and no SAEs reported
- No AEs leading to death

Pharmacokinetics

Individual CEBPA-51 plasma concentration vs time profiles after co-administration of either A) 70 (n=4), B) 98 (n=3) or C) 130 (n=1) mg/m² MTL-CEBPA on D1 and D8 and 200 mg pembrolizumab on D2 (indicated by an arrow). Plasma CEBPA-51 concentration vs time profiles were collected over 7 days after the first dose (D1) and over 3 days after the second dose (D8).

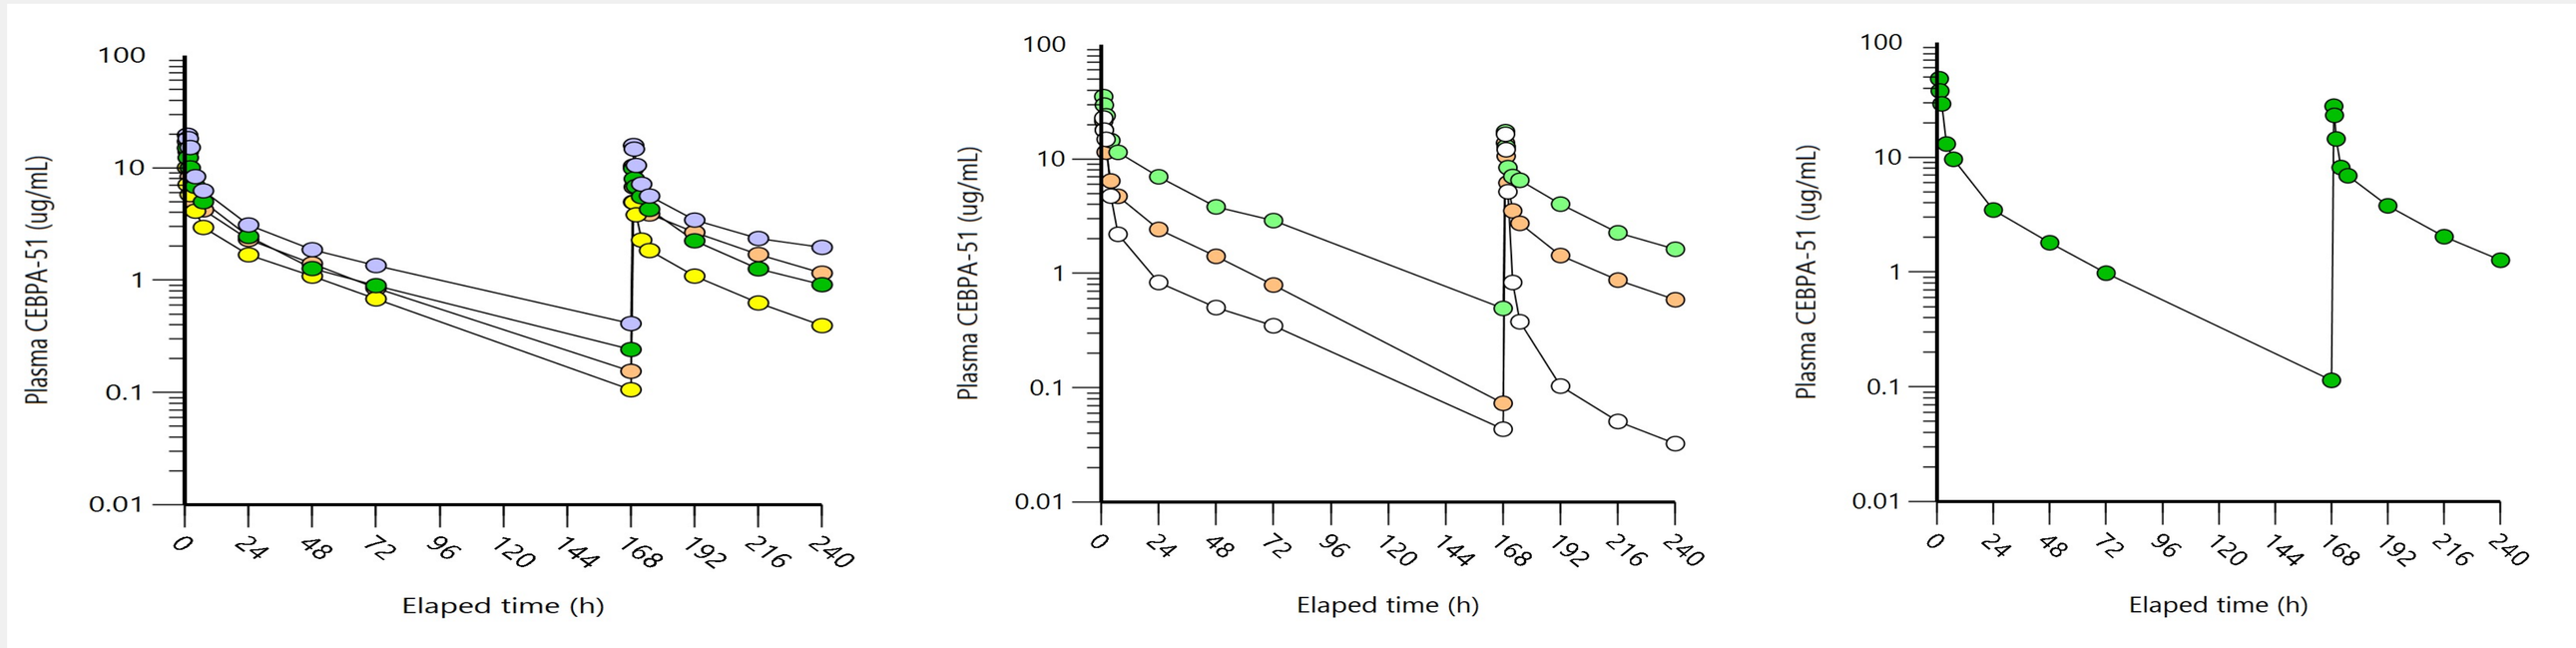
Plasma CEBPA-51 (ug/mL)
Elaped time (h)

Treatment Outcomes

A) Swimmer plot for all evaluable patients in study

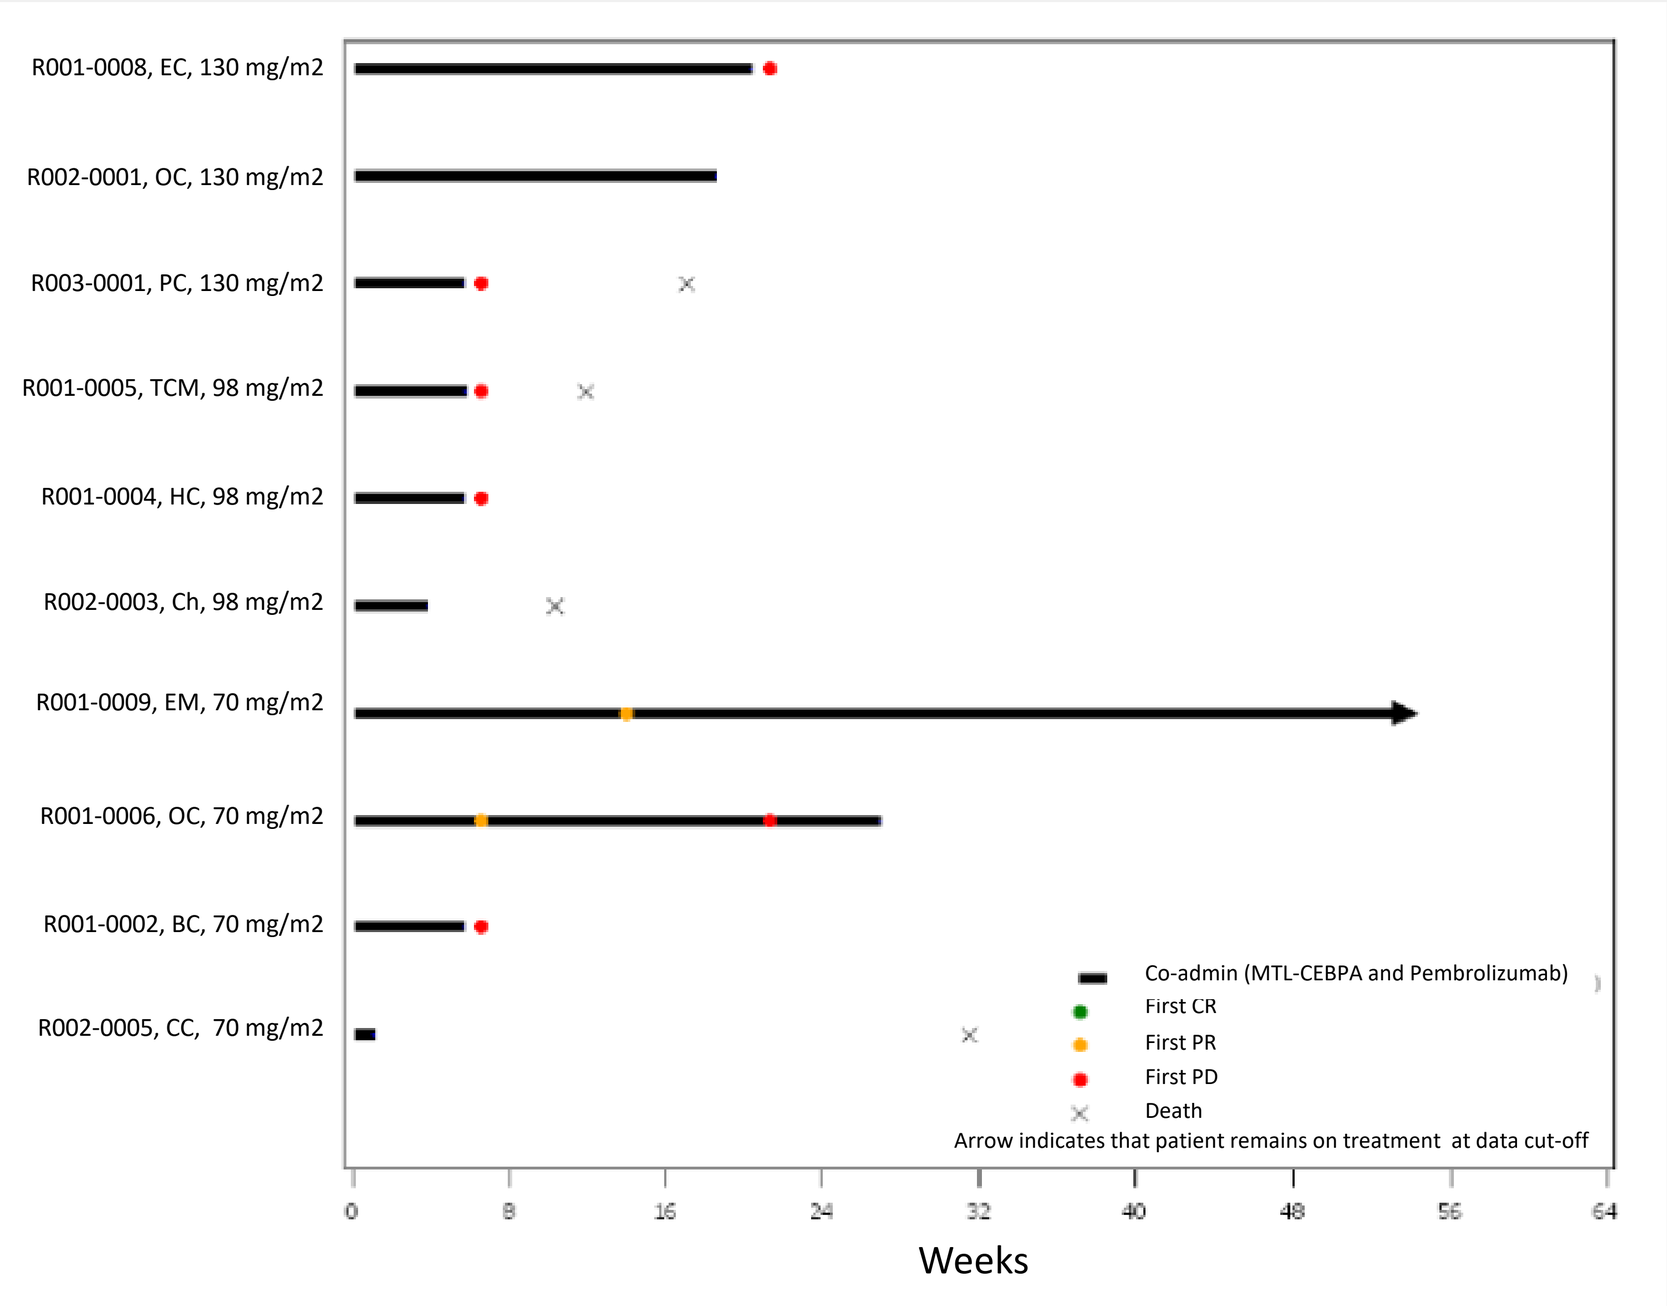
R001-0008, EC, 130 mg/m2
R002-0001, OC, 130 mg/m2
R003-0001, PC, 130 mg/m2
R001-0005, TCM, 98 mg/m2
R001-0004, HC, 98 mg/m2
R002-0003, Ch, 98 mg/m2
R001-0009, EM, 70 mg/m2
R001-0006, OC, 70 mg/m2
R001-0002, BC, 70 mg/m2
R002-0005, CC, 70 mg/m2
Co-admin (MTL-CEBPA and Pembrolizumab)
First CR
First PR
First PD
Death
Arrow indicates that patient remains on treatment at data cut-off
Weeks

B) Best change in tumour size by primary tumour

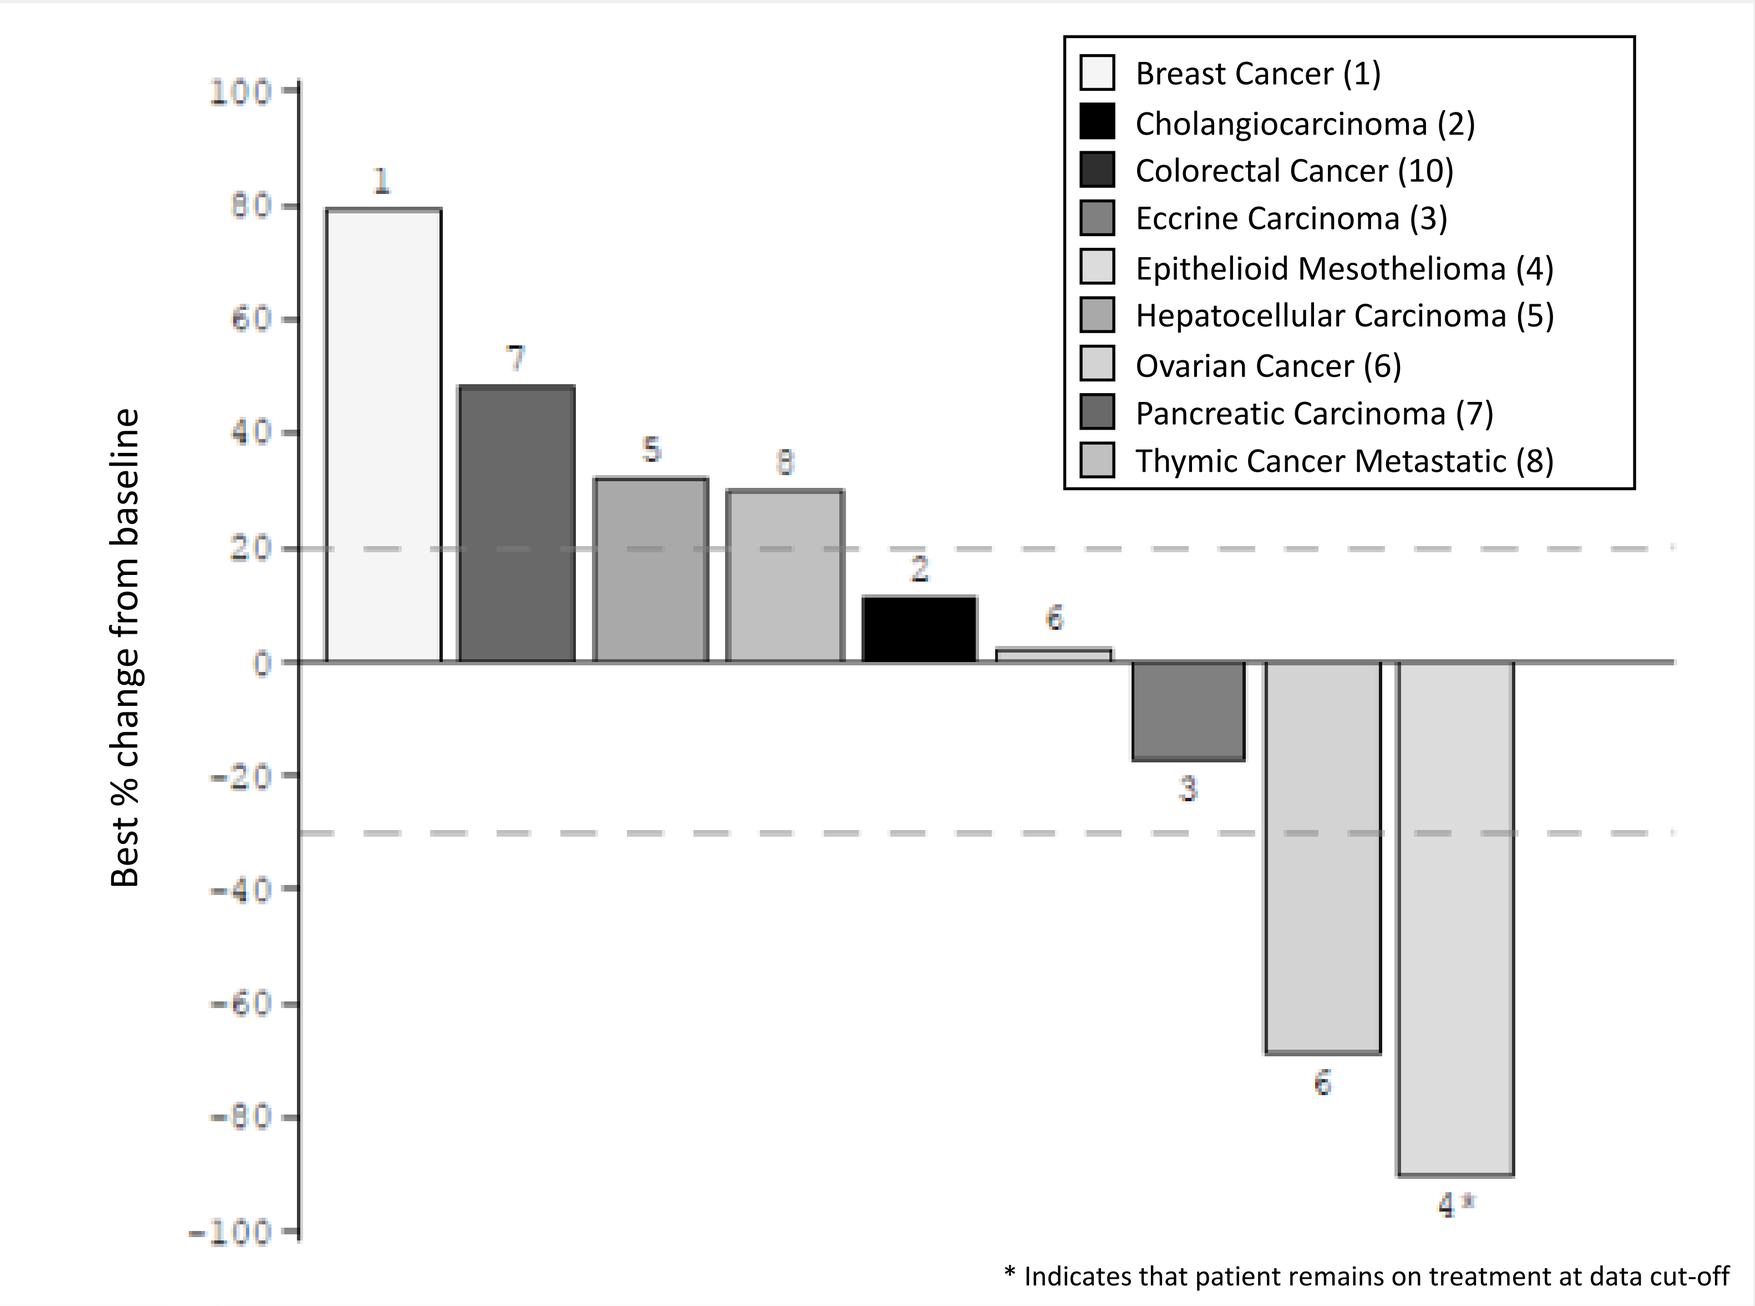
Breast Cancer (1)
Cholangiocarcinoma (2)
Colorectal Cancer (10)
Eccrine Carcinoma (3)
Epithelioid Mesothelioma (4)
Hepatocellular Carcinoma (5)
Ovarian Cancer (6)
Pancreatic Carcinoma (7)
Thymic Cancer Metastatic (8)
Best % change from baseline
* Indicates that patient remains on treatment at data cut-off

C) Spider plot illustrating each patient and primary tumour

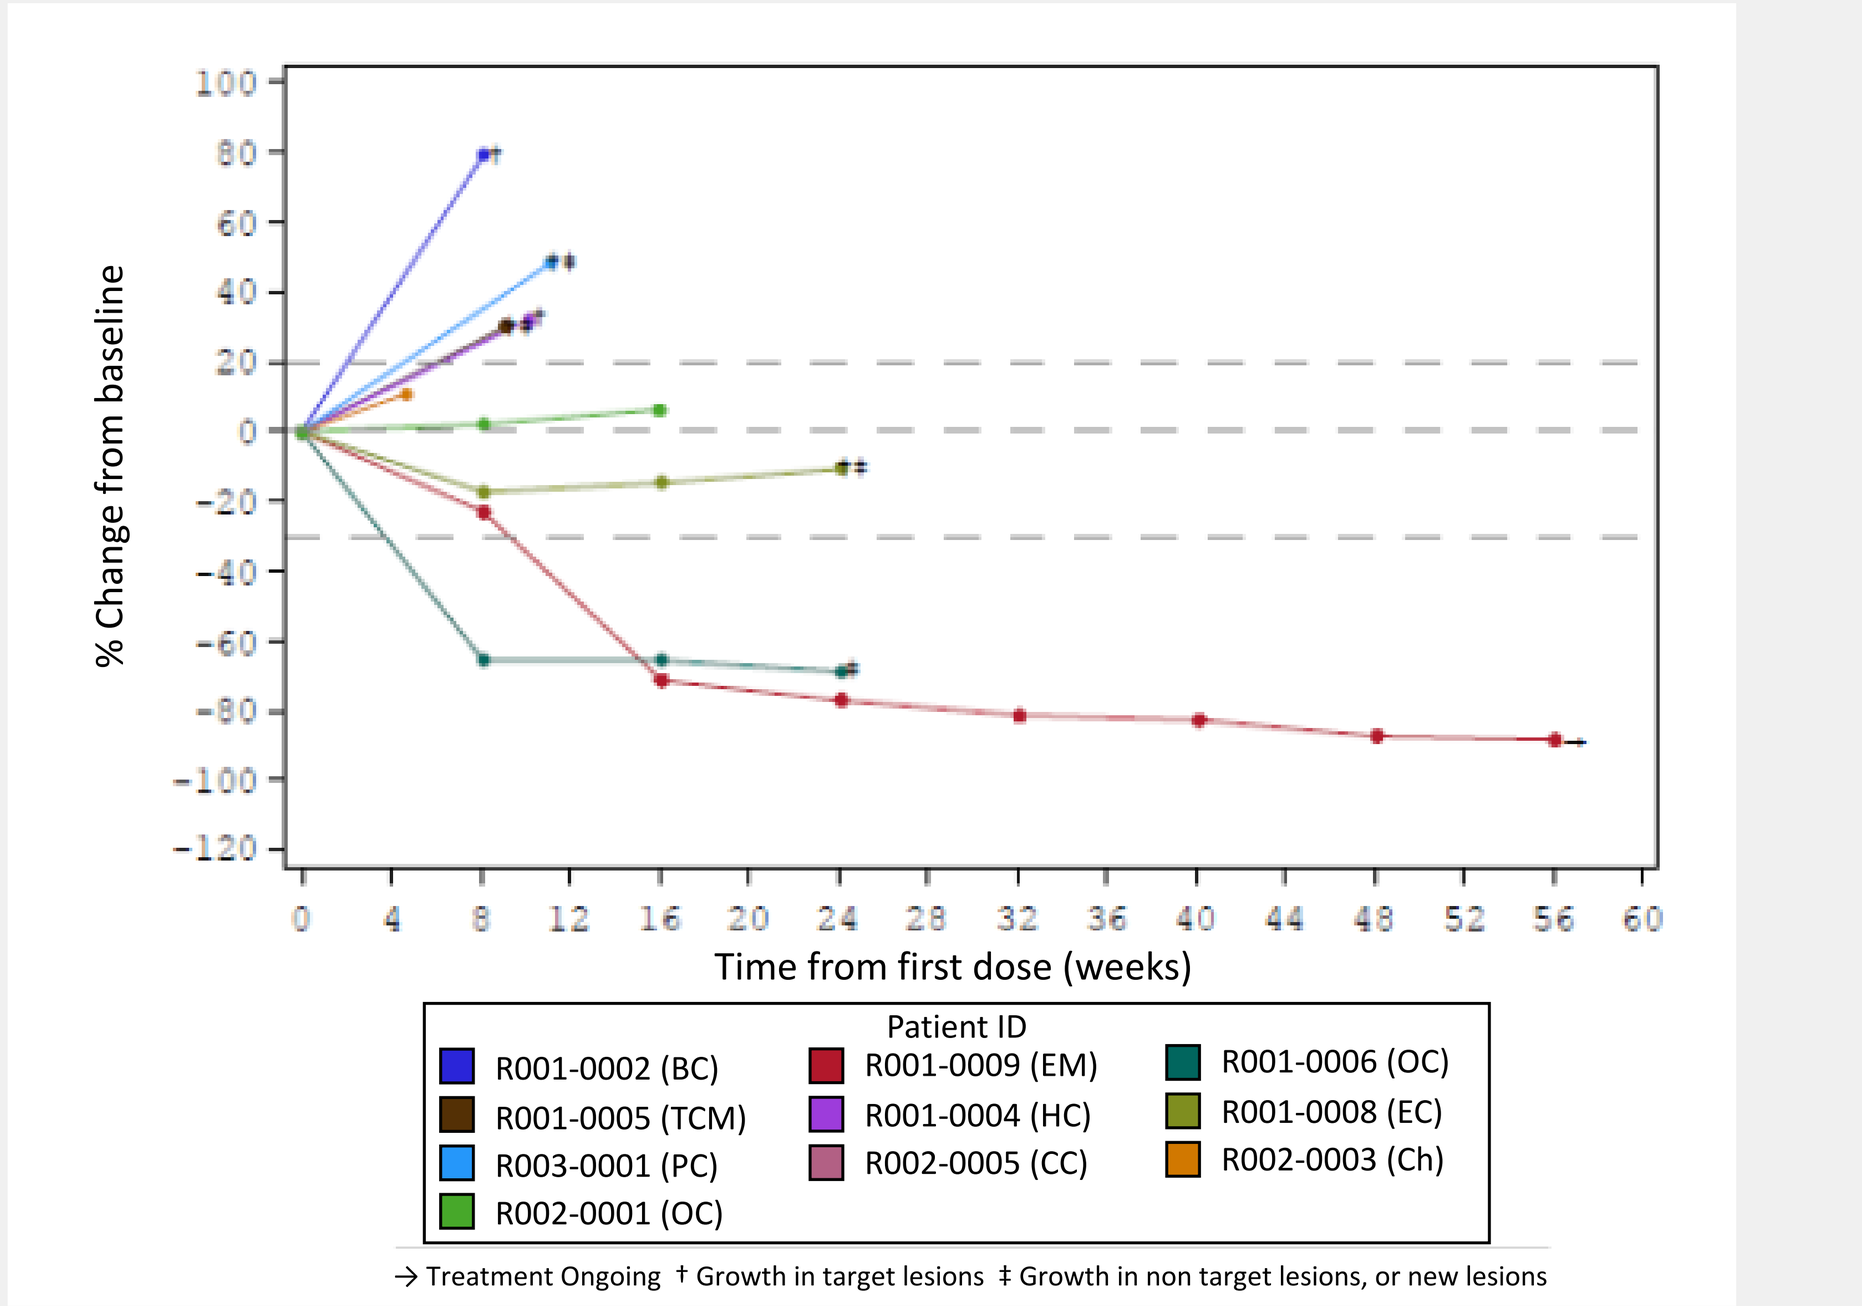
% Change from baseline
Time from first dose (weeks)
Patient ID
R001-0002 (BC)
R001-0005 (TCM)
R003-0001 (PC)
R002-0001 (OC)
R001-0009 (EM)
R001-0004 (HC)
R002-0005 (CC)
R001-0006 (OC)
R001-0008 (EC)
R002-0003 (Ch)
→ Treatment Ongoing † Growth in target lesions ‡ Growth in non target lesions, or new lesions

BC = Breast Cancer; EM = Epithelioid Mesothelioma; OC = Ovarian Cancer; TCM = Thymic Cancer Metastatic; HC = Hepatocellular Carcinoma; EC = Eccrine Carcinoma; PC = Pancreatic Carcinoma; CC = Colorectal Cancer; Ch = Cholangiocarcinoma

Partial Responders

The first case is a 56 year old female diagnosed in March 2018 with epithelioid mesothelioma of the left pleura (stage IV – T3N2M0). Her prior treatments included carboplatin, cisplatin and pemetrexed chemotherapy. Her tumour sites on enrolment in the study were mediastinal lymph nodes, muscle, soft tissue and pleura. Following 2 months of treatment in the 70mg/m2 cohort she was observed to have 23% tumour reduction and this response has increased to 83% tumour reduction and is ongoing at 9 months. Below, a target mediastinal lymph node lesion shown at baseline, C2 day 22 and C10 day 22:

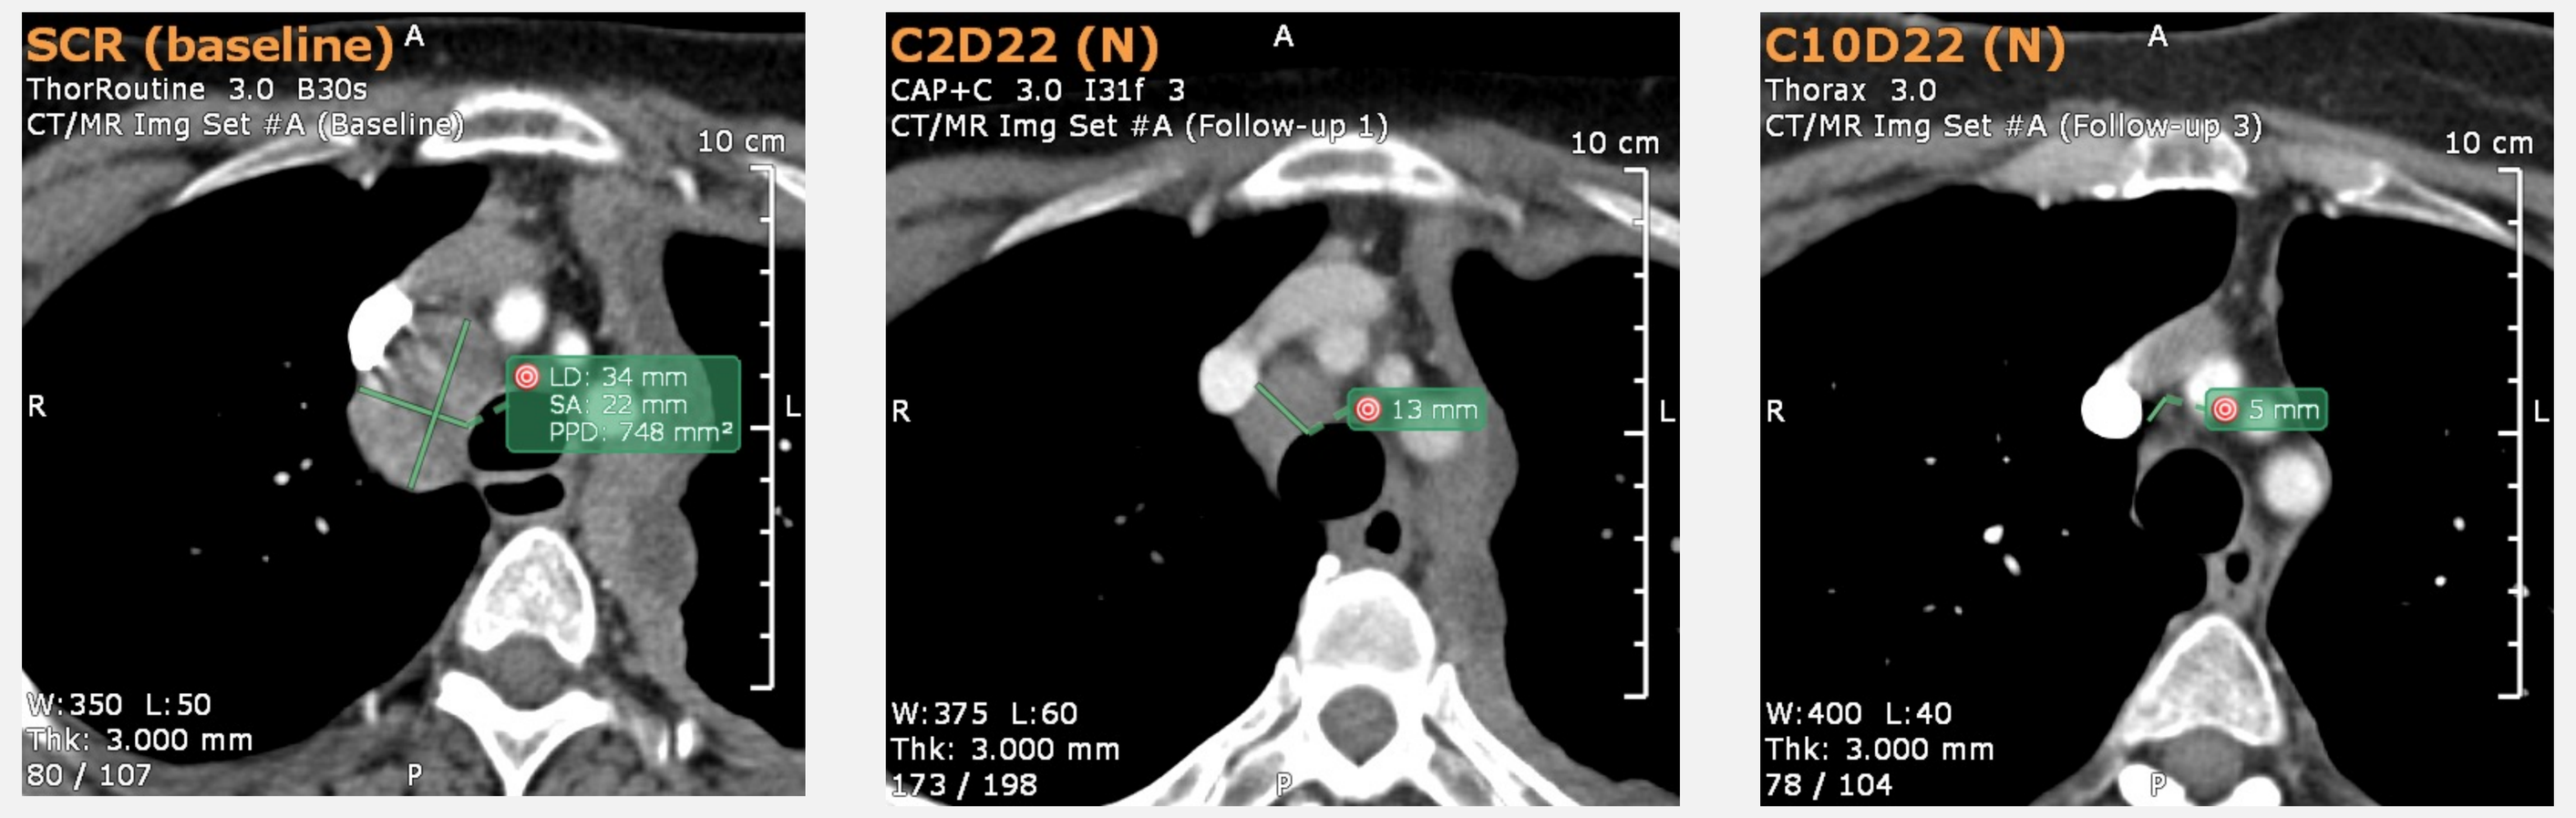
SCR (baseline)
C2D22 (N)
C10D22 (N)
13 mm
5 mm

The second case is a 53 year old female with a primary diagnosis of high grade serous ovarian cancer (HGSOC) for which she has undergone surgery in the form of hysterectomy, omentectomy, colectomy, radical hysterectomy, bilateral salpingoophorectomy and splenectomy in July 2012. On entering the trial she had progressed on 6 previous lines of systemic therapy included bevazicumab, carboplatin, doxorubicin, gemcitabine, olaparib and paclitaxel. Following 2 months of treatment in the 70mg/m2 cohort she was noted to have a 66% decrease in tumour volume which increased to a maximum of 69% before she was withdrawn due disease progression at the end of cycle 6.

Below, a target retroperitoneal lymph node lesion shown at baseline and cycle 2 day 22. The patient had her target lesions disappeared at next scan (maintained thereafter), with disease progression in cycle 6 due to new lesions:

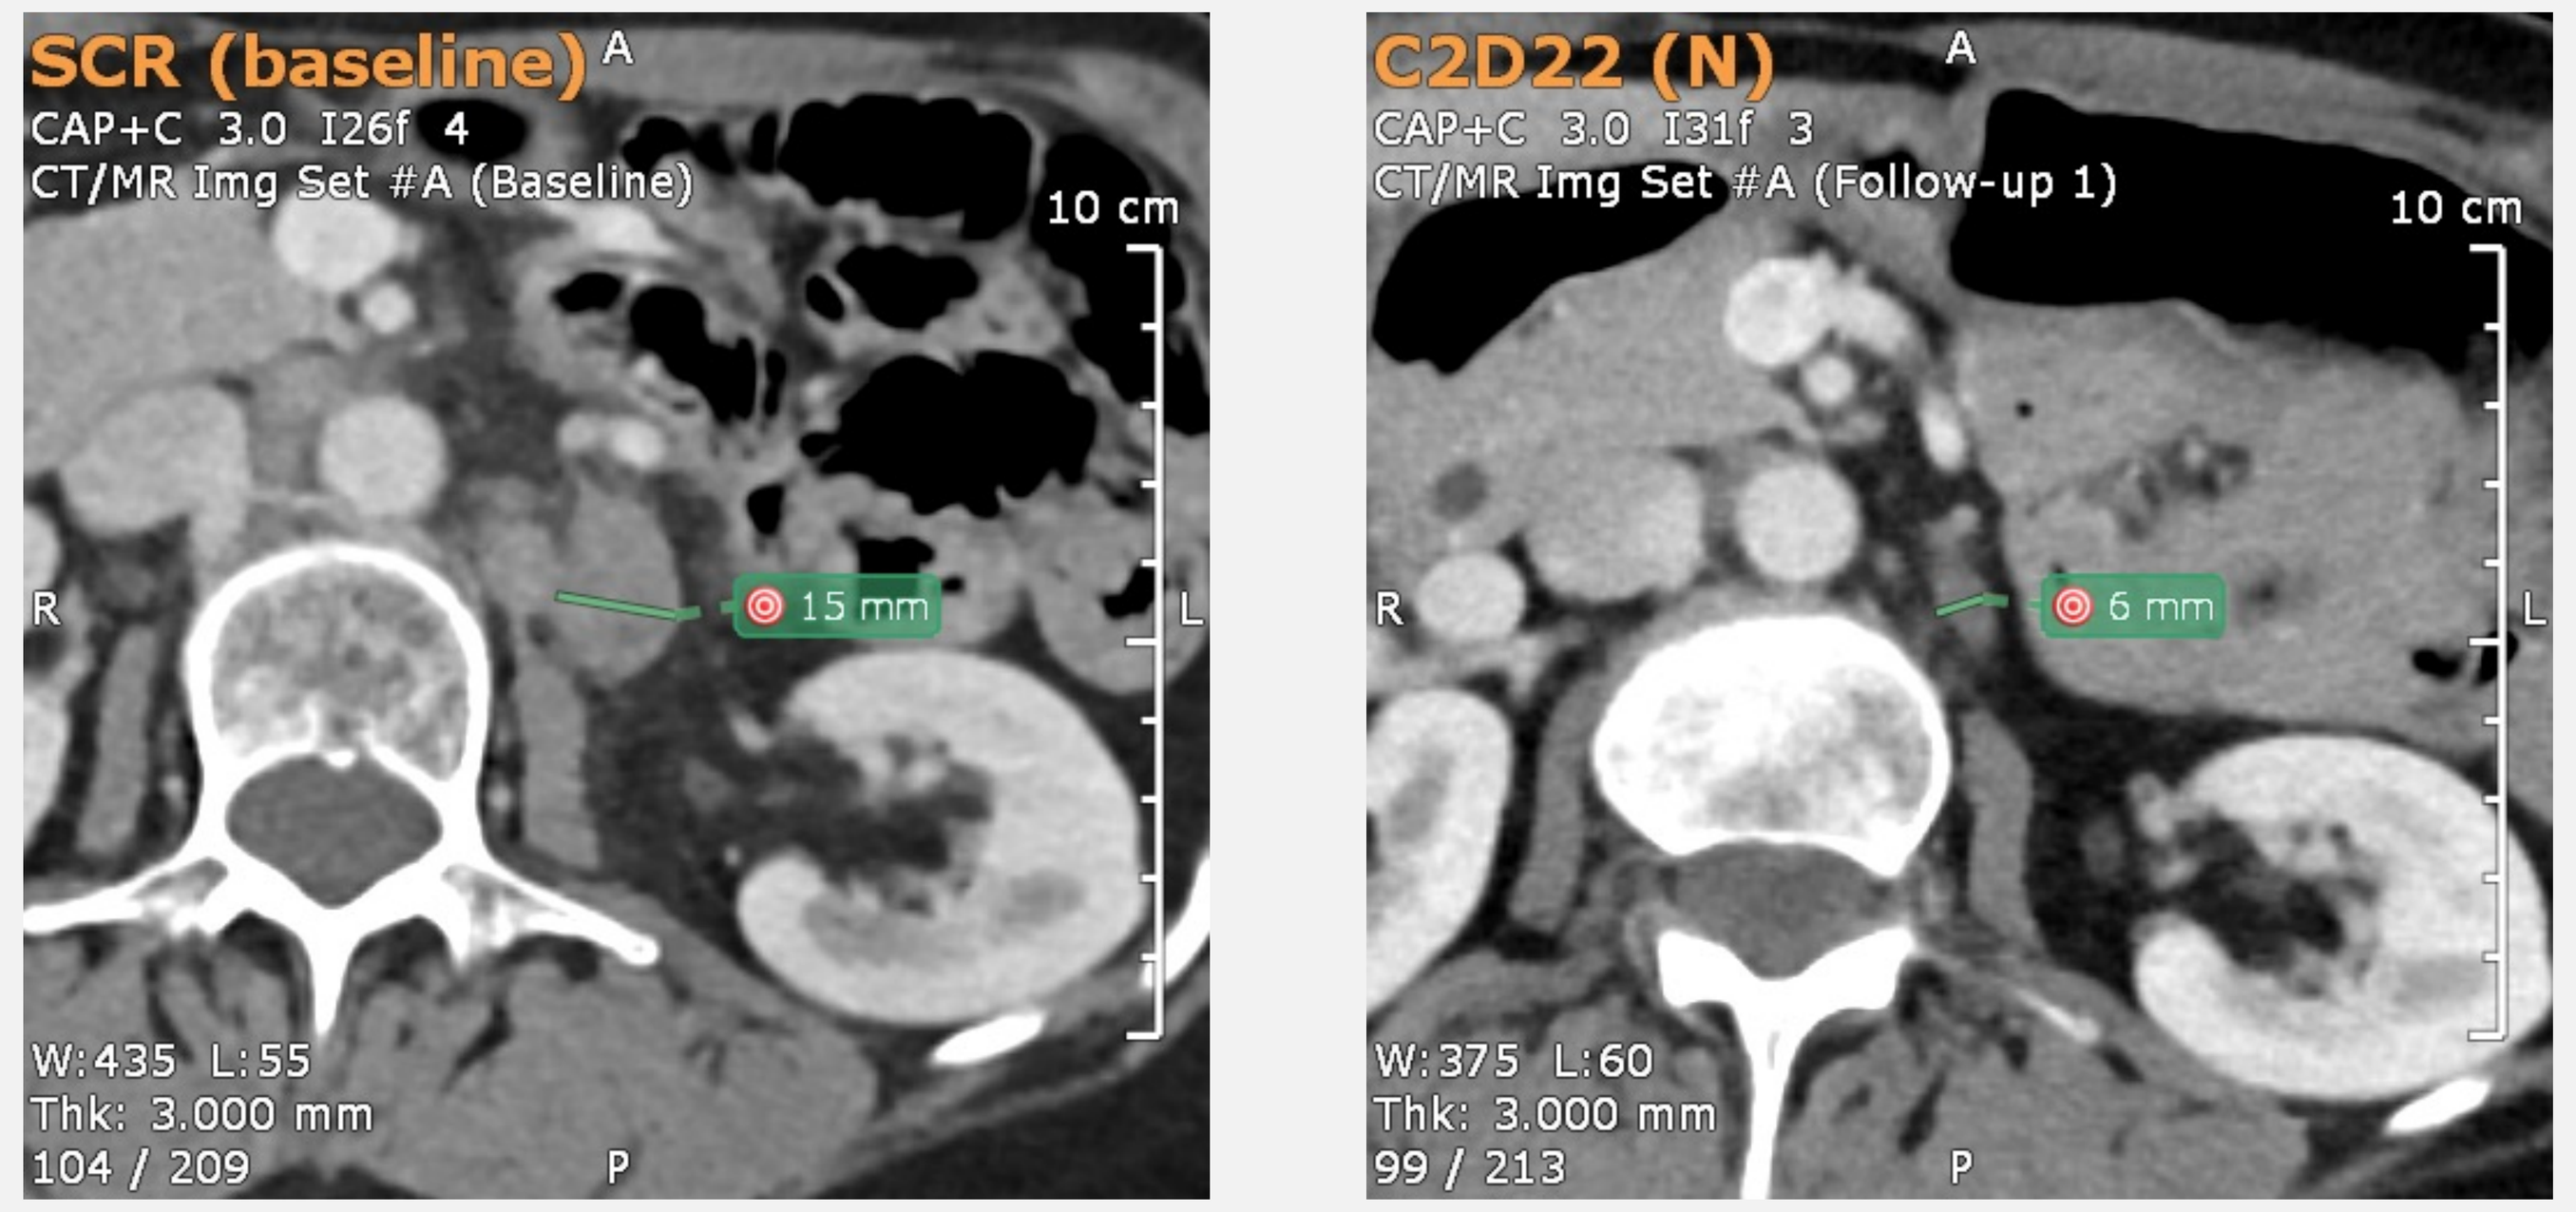
SCR (baseline)
C2D22 (N)
15 mm
6 mm

The second patient recruited with HGSOC was a heavily pre-treated 40 yr old, with 6 prior lines of systemic therapy, who achieved SD (max tumor shrinkage -24%).

The study is conducted with blinded independent central radiology review in place. Concordance so far is high with no differences in the assessment of BOR between local and central review. All above images are from the central review reads.

Conclusion

MTL-CEBPA in combination with pembrolizumab demonstrated manageable toxicity at the dose levels tested and has shown antitumor activity. MTD was not reached and RP2D was determined at 130mg/m2 on day 1, 8 and 15 of a 28 day cycle. Enrolment into the dose expansion is ongoing.